ABSTRACT
#C204

CHARACTERIZATION OF TP53 MUTATIONS BY DNA AND RNA SEQUENCING OF HIGH GRADE SEROUS OVARIAN CANCER (HGSOC) FFPE SECTIONS

Diane Ilsley[1], Marie Fahey[1], Brian C Haynes[1], Gary J Latham[1], Huiping Zhu[1], Andrew Kincaid[1], Jeffrey Shelton[1], Elizabeth B Somers[2], and Daniel J O'Shannessy[2]

[1]Asuragen, Inc., Austin, TX; [2]Morphotek, Inc., Exton, PA

SUMMARY

- Primary FFPE tumor samples from a cohort of >200 high grade serous ovarian cancer (HGSOC) patients were profiled using an integrated analysis comprised of both DNA and RNA sequencing to gain a more complete understanding of the functional consequences of *TP53* mutations.
- *TP53* mutation status was determined using the QuantideX® NGS *TP53* Coding Exon Panel and the AmpliSeq® Cancer Hotspot Panel, and regions of *TP53* covered by both assays were found to be concordant.
- Mutation status determined using matched RNA-Seq data was found to be highly concordant with the QuantideX® NGS panel.
- RNA-Seq gene expression data was integrated with the DNA mutation data resulting in the identification of a set of high confidence associations between *TP53* mutations and RNA expression changes in genes regulated by p53.

INTRODUCTION

TP53 is the most frequently mutated gene across all cancers at approximately 50%, and in cancers such as HGSOC, up to 96%. In addition to the high incidence of DNA mutations at the gene locus, the *TP53* pathway is also dysregulated through alternative mechanisms which involve other members of the regulatory cascade, such as overexpression of MDM2. Asuragen has developed sensitive NGS assays and algorithms to characterize DNA mutations and RNA expression profiles from the most challenging clinical samples, such as archival FFPE samples. In order to enable a more complete understanding of the functional consequences of *TP53* mutations (or lack thereof), we present an application of these approaches to characterize *TP53* function in FFPE sections from HGSOC through an integrated analysis of DNA and RNA sequencing.

METHODS

Primary debulking surgery FFPE tumor sections were collected from a cohort of 235 HGSOC patients. DNA from macro-dissected tumor and matched normal blood specimens were profiled by the QuantideX® NGS *TP53* Coding Exon Assay (Asuragen, Inc.), and library analysis was performed using the QuantideX® NGS Reporter. *TP53* mutation status of the tumor specimens was independently assessed by the AmpliSeq Cancer Hotspot Panel v1 (Thermo Fisher Scientific, Inc.). Whole transcriptome RNA-Seq was performed using ribosomal RNA-depleted total RNA, strand-specific libraries, and 50 bp paired-end sequencing. Gene and isoform expression quantitation as well as SNV and indel calling was performed on the RNA-Seq libraries.

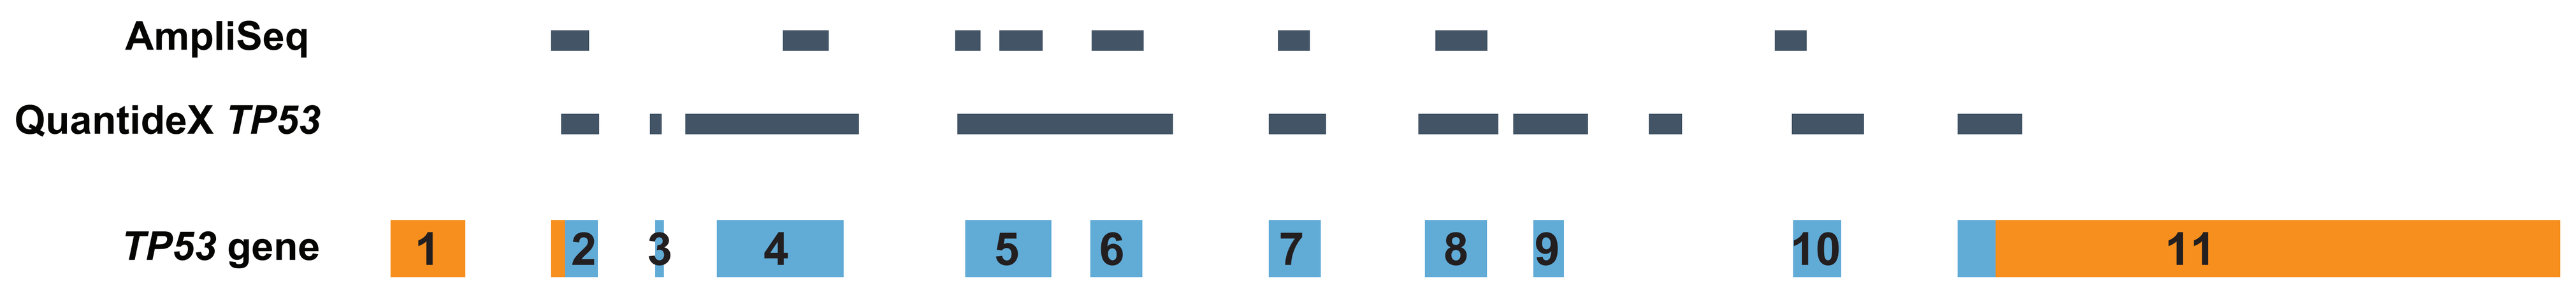

AmpliSeq
QuantideX TP53
TP53 gene
1
2
3
4
5
6
7
8
9
10
11

Figure 1. QuantideX and AmpliSeq NGS panel gene coverage of *TP53*. The coding exons are represented in blue and the noncoding exons in orange. The QuantideX *TP53* panel covers 100% of the *TP53* coding bases, 99.5% of all COSMIC variants and all ovarian TCGA *TP53* variants (390). In contrast, the AmpliSeq Hotspot panel covers 68.4% of the coding bases, 68.4% of the *TP53* COSMIC variants and 303 ovarian TCGA variants.

*Research Use Only – Not For Use In Diagnostic Procedures
Preliminary research data. The performance characteristics of this assay have not yet been established.
Presented at EORTC 2015

RESULTS

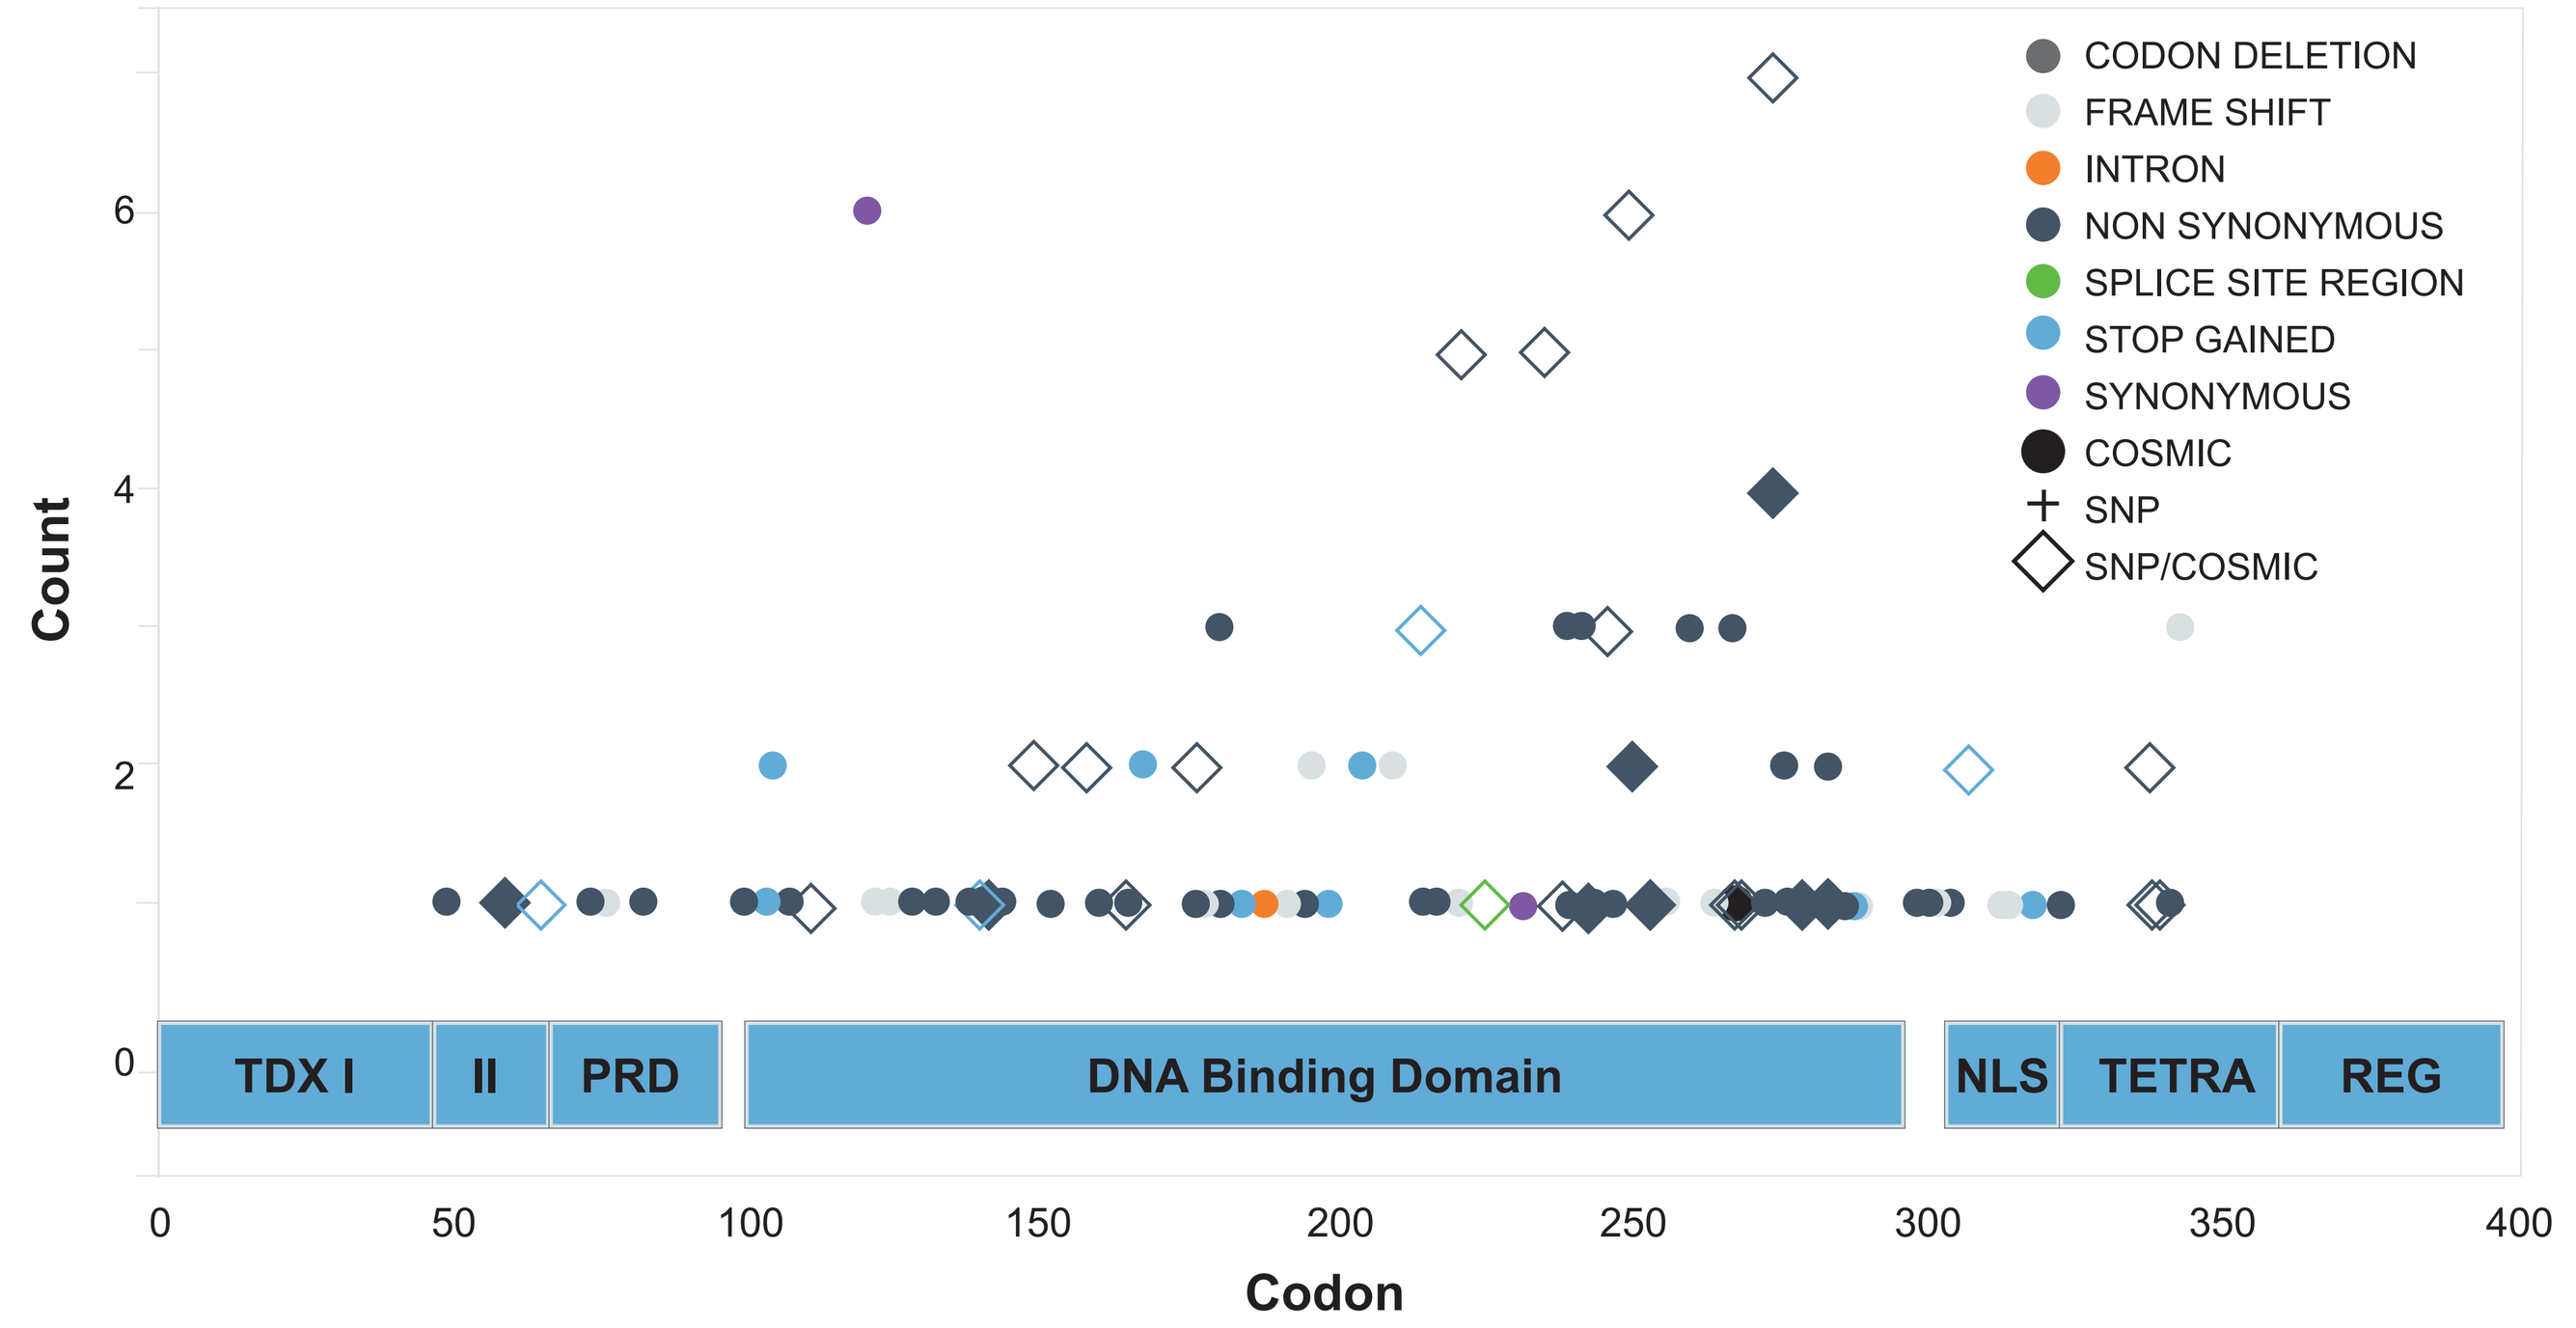

CODON DELETION
FRAME SHIFT
INTRON
NON SYNONYMOUS
SPLICE SITE REGION
STOP GAINED
SYNONYMOUS
COSMIC
SNP
SNP/COSMIC
Count
0
2
4
6
TDX I
II
PRD
DNA Binding Domain
NLS
TETRA
REG
0
50
100
150
200
250
300
350
400
Codon

Figure 2. Variants identified by the QuantideX NGS panel aligned to the p53 protein domains. The plot shows the distribution of both SNVs (187 somatic) and indels (37), which were found to be mutually exclusive at the patient level. Consistent with previous studies, *TP53* mutations were present in 84% of the HGSOC cohort. Symbol color indicates the type of mutation and symbol shape indicates variant annotation.

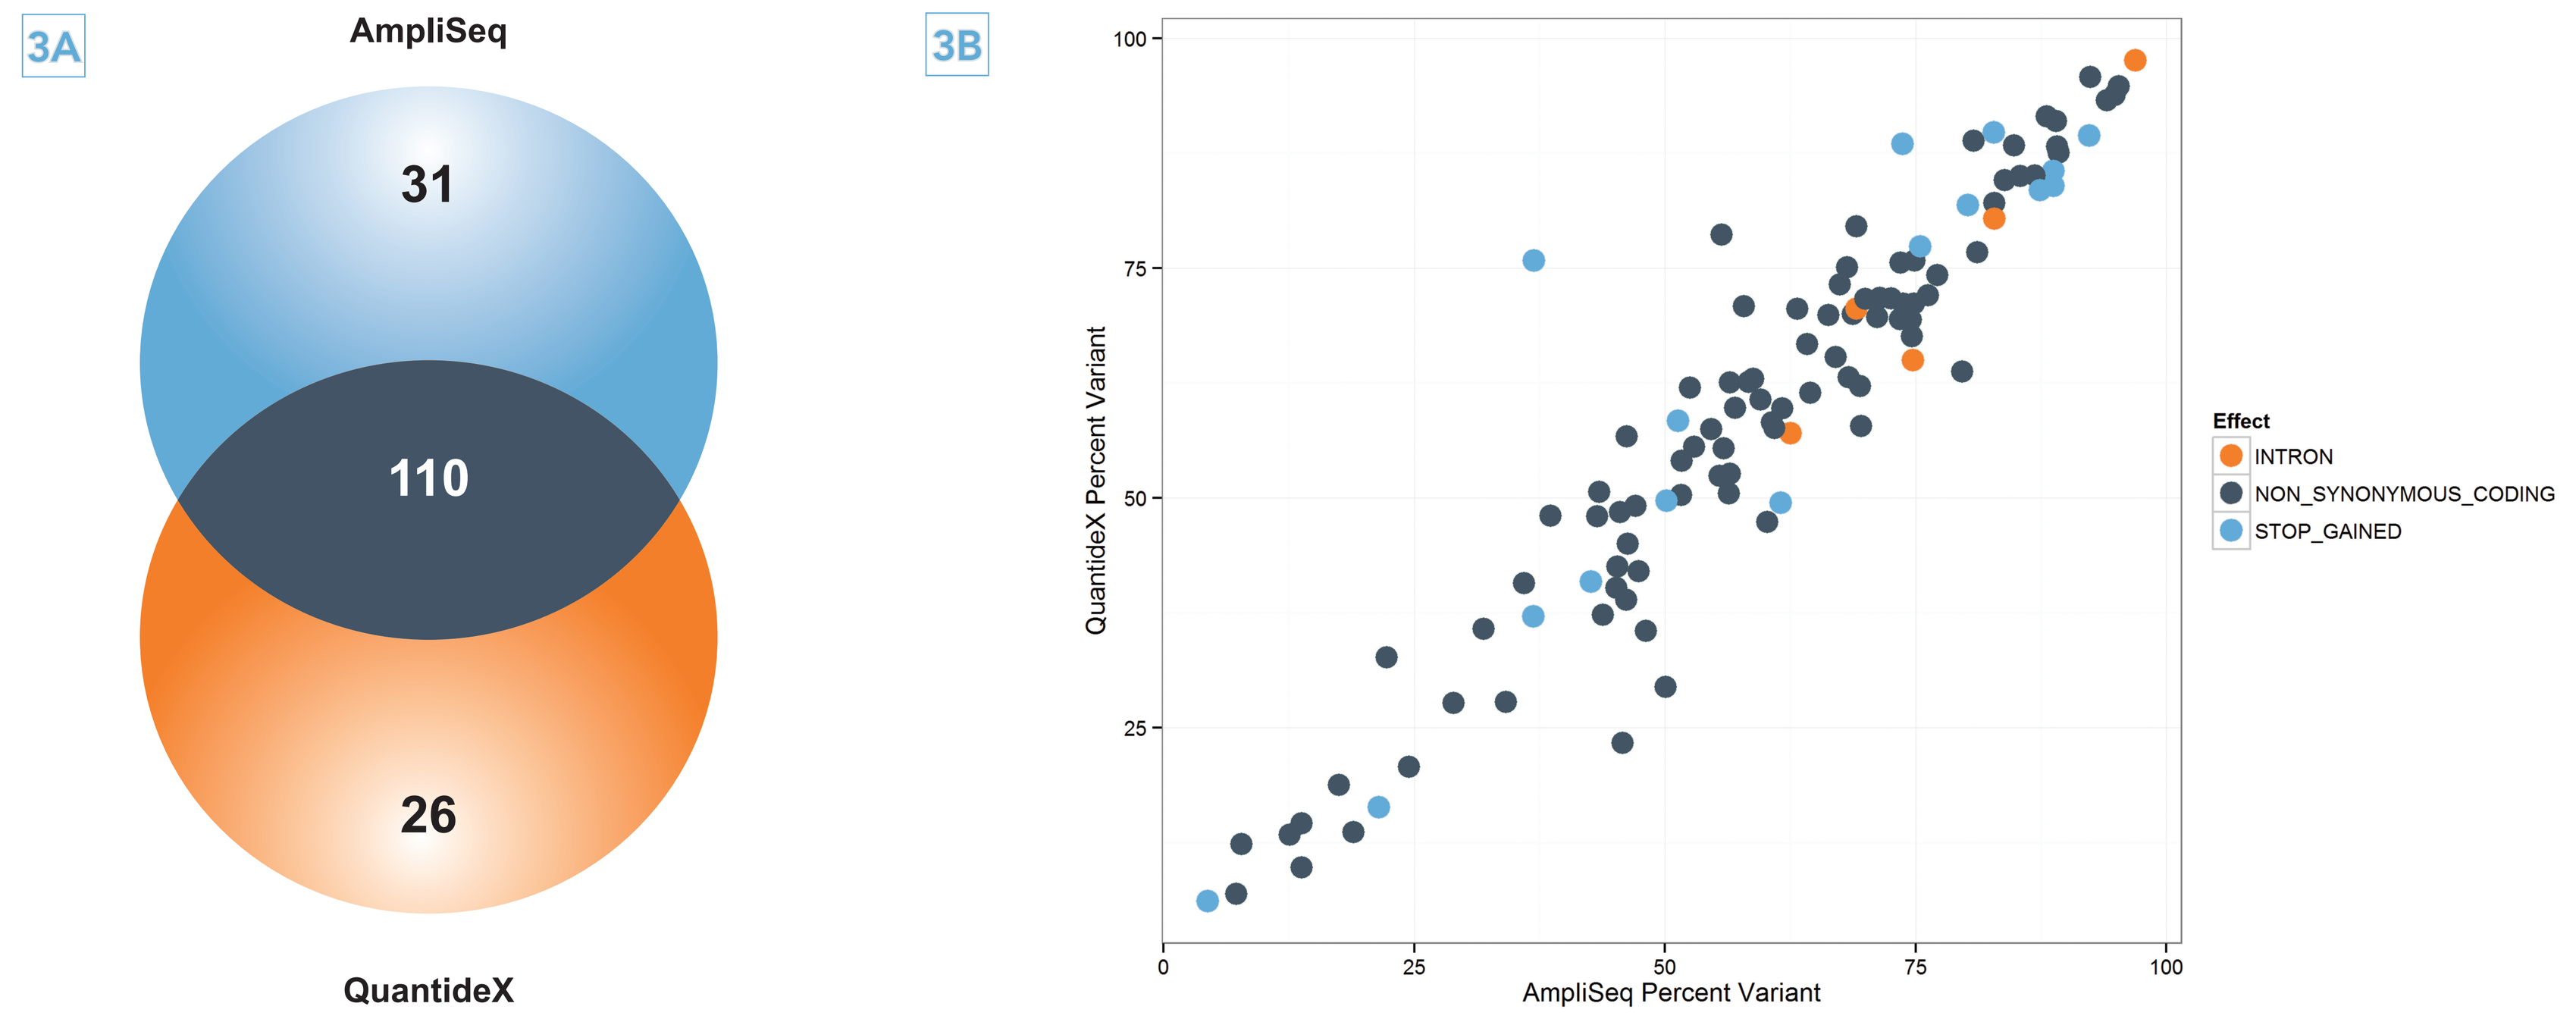

3A
AmpliSeq
31
110
26
QuantideX
3B
QuantideX Percent Variant
AmpliSeq Percent Variant
0
25
50
75
100
Effect
INTRON
NON_SYNONYMOUS_CODING
STOP_GAINED

Figure 3. Positive call summary comparing results by panel. A) The Venn diagram shows the concordance of variant calls in gene regions covered by both QuantideX and AmpliSeq after filtering for germline variants and applying a coverage filter (100X). RNA-Seq data confirmed 6/26 positive calls in QuantideX and 2/31 positive calls in AmpliSeq. The variants called positive by AmpliSeq (29/31) were not called positive by QuantideX because they did not meet one of the following criteria: sensitivity threshold, coverage filter, or contain matching forward and reverse reads. Considering the total content coverage of the QuantideX Panel, QuantideX called an additional 51 variants that were not covered by the AmpliSeq panel. **B)** Variant frequency comparison of QuantideX (Y-axis) versus AmpliSeq (X-axis) for the 110 variants called by both panels. Variants are colored by mutation functional class.

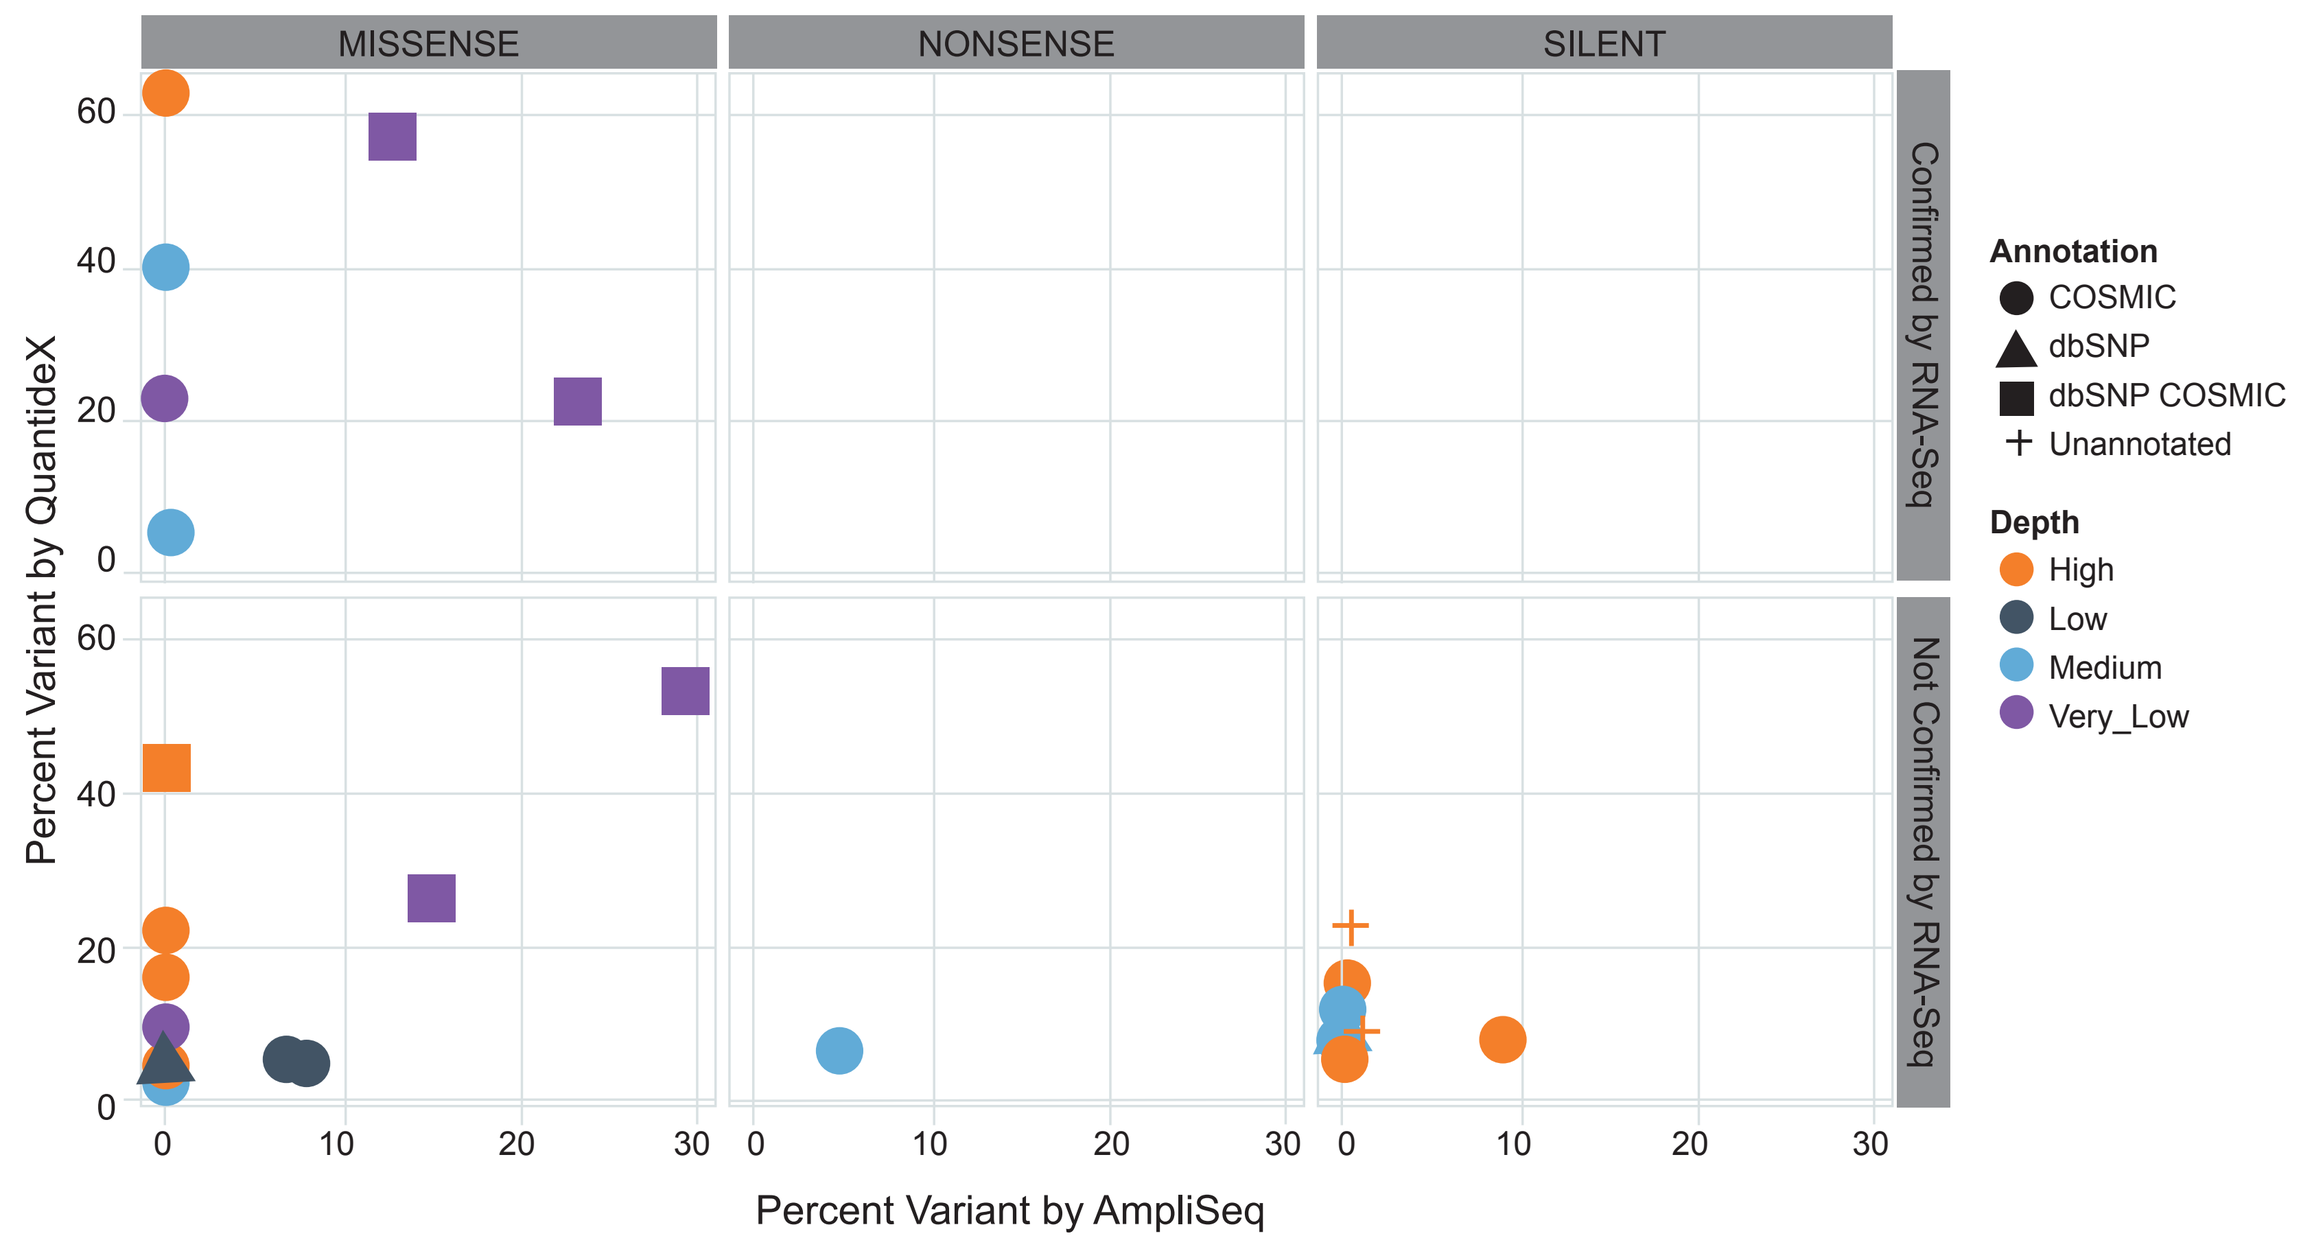

MISSENSE
NONSENSE
SILENT
Confirmed by RNA-Seq
Not Confirmed by RNA-Seq
Percent Variant by QuantideX
Percent Variant by AmpliSeq
0
10
20
30
40
60
Annotation
COSMIC
dbSNP
dbSNP COSMIC
Unannotated
Depth
High
Low
Medium
Very_Low

Figure 4. QuantideX identifies positive variants missed by AmpliSeq. The variant frequency by QuantideX (Y-axis) is plotted versus the variant frequency by AmpliSeq (X-axis) highlighting the variant calls missed by AmpliSeq due low sequencing coverage (very low = <100X) or below the sensitivity threshold. The symbol shape is variant annotation and symbol color reflects depth of sequencing coverage in AmpliSeq. The top panels show the variants that were confirmed by RNA-Seq while the bottom panel contains variants not confirmed by RNA-Seq due to low sequencing coverage (2-20X).

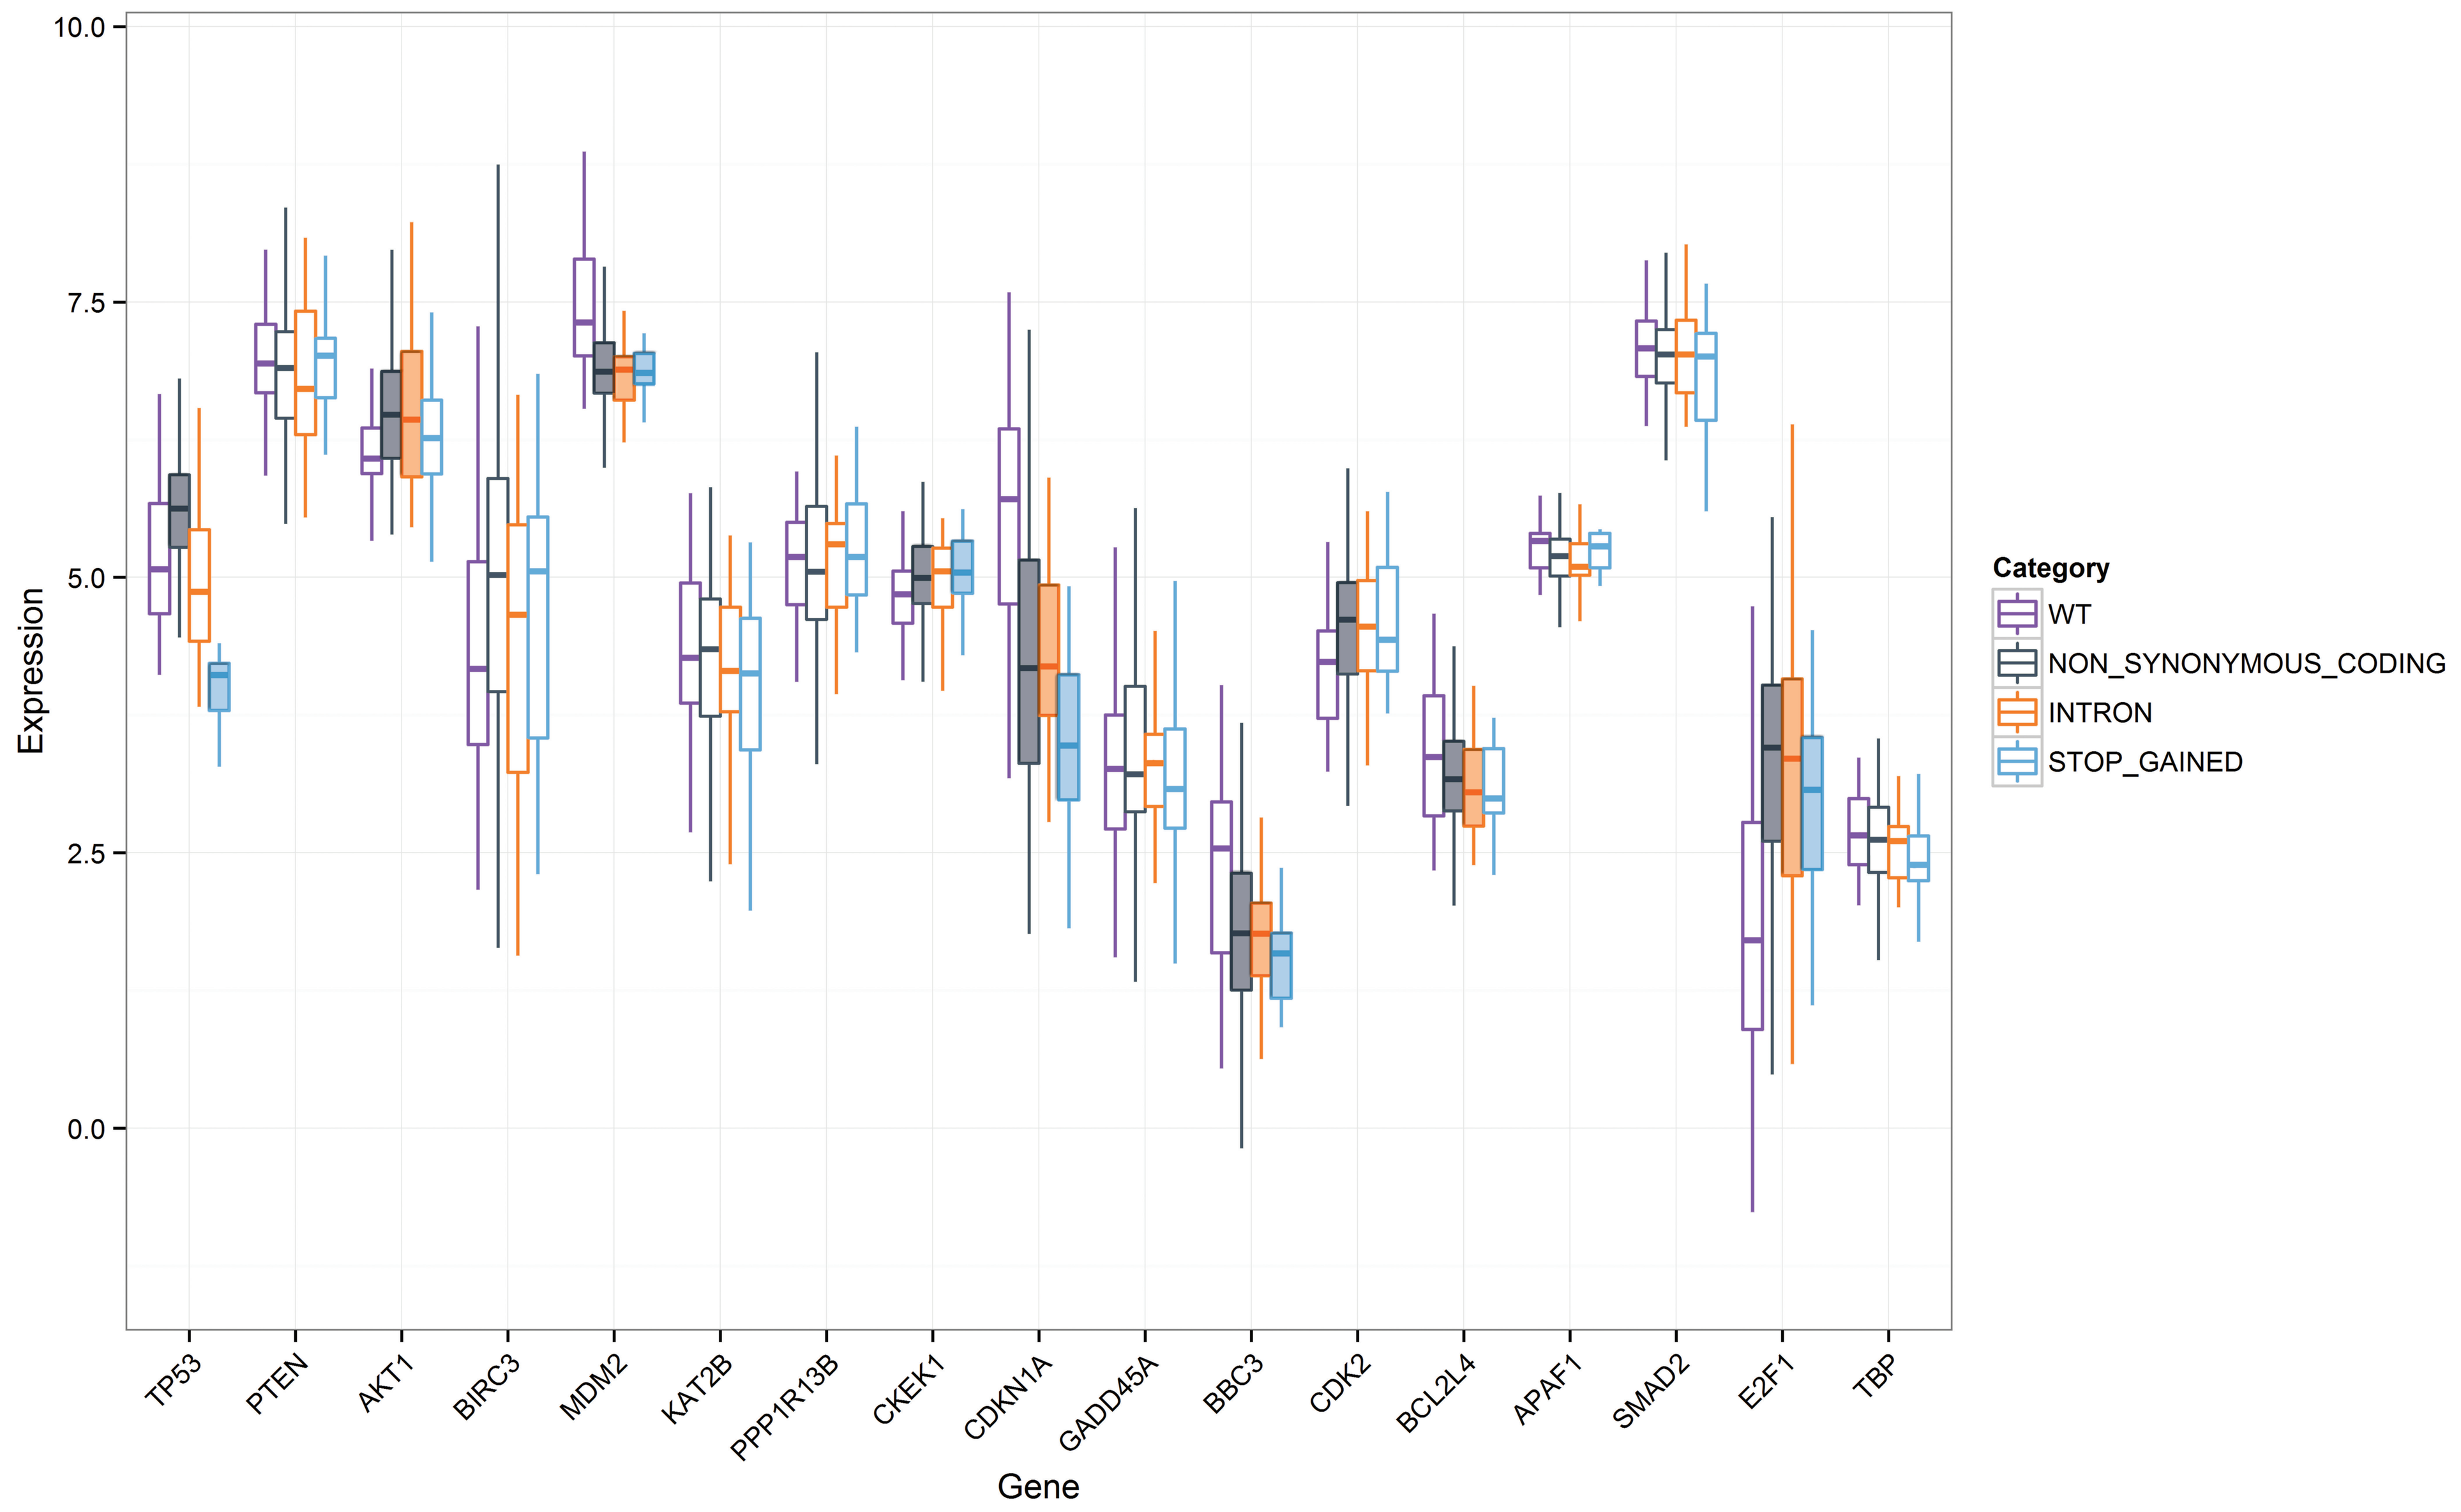

Expression
0.0
2.5
5.0
7.5
10.0
TP53
PTEN
AKT1
BIRC3
MDM2
KAT2B
PPP1R13B
CKEK1
CDKN1A
GADD45A
BBC3
CDK2
BCL2L4
APAF1
SMAD2
E2F1
TBP
Gene
Category
WT
NON_SYNONYMOUS_CODING
INTRON
STOP_GAINED

Figure 5. *TP53* variant type differentially affects *TP53* expression and expression of *TP53* regulated genes. The graph shows boxplots of the median log2 CPM for a subset of genes (indicated along the X-axis) directly or indirectly regulated by p53. *TP53* variant type is indicated by color and boxplots shaded highlight the genes and *TP53* variants that are differentially expressed relative to wild-type *TP53* based on a FDR corrected p-value < 0.05.

CONCLUSIONS

- We successfully used NGS to characterize the DNA mutations and RNA expression profiles from a cohort of 235 archival HGSOC FFPE specimens.
- Because *TP53* is highly mutated across all exons, a hot spot mutation panel does not provide adequate gene coverage for mutation assessment. The QuantideX® NGS *TP53* Panel identified mutations in 60 additional patients that would have been missed by a hotspot panel.
- Integration of *TP53* DNA mutation data with matching RNA-Seq data provides a more complete understanding of the functional consequences of the different classes of *TP53* mutations through association of gene signatures with gain and loss of p53 function.

morphotek®

Asuragen®
www.asuragen.com